Phase 1/2a trial of CD200R1 inhibitor 23ME-00610: exploratory analyses of tissue-based and genetic biomarkers

A.R. RAZAK,[1*] A. NAING,[2] A. DIEP,[3] C.C. HWANG,[3] C. HOM,[3] P. SOOD,[3] A. REISETTER,[3] P. NANDAKUMAR,[3] Z. FULLER,[3] D. GLATT,[3] M. SCHMIDT[3]

[1]Princess Margaret Cancer Centre, University of Toronto, Toronto, CA [2]The University of Texas MD Anderson Cancer Center, Houston, TX, USA [3]23andMe South San Francisco, CA, USA *corresponding

23andMe Therapeutics

FPN # 150P

ESMO 2024 Annual Meeting
September 13-17, 2024
Barcelona, Spain

Copies of this poster obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

Genetic Signature

- Using the 23andMe database, pleiotropic causal variants with opposing effect on risks for cancer and immune diseases, referred to as an immuno-oncology signature, were observed in 3 components of the CD200R1 pathway, including CD200R1, its ligand CD200 and the signaling adaptor protein DOK2.[1] **(Figure 1)**

CD200R1

- CD200R1 is expressed on immune cells and binds to CD200, downregulating proinflammatory cytokines by activated T and myeloid cells and promoting an immunosuppressive microenvironment in cancers, where CD200 is highly expressed.[1] **(Figure 2)**

23ME-00610

- 23ME-00610 is a first-in-class IgG1 antibody that binds CD200R1 with high affinity (K_D < 0.1 nM) and inhibits immunosuppressive signaling, leading to restoration of T cell activity and killing of CD200-expressing tumor cells in preclinical studies.[1]
- 23ME-00610 is being evaluated in a Phase 1/2a clinical trial in patients with advanced solid cancers **(NCT05199272) (Figure 3)** and has demonstrated acceptable safety and tolerability and anti-tumor activity in a neuroendocrine tumor patient and a renal cell carcinoma patient.[2]
- For the first time, the exploratory tissue-based biomarker data and genetic analysis are presented.

Figure 1. CD200R1 Pathway Identified as a Critical Immune Checkpoint with 23andME's Immuno-oncology (I/O) Genetic Signature[1]

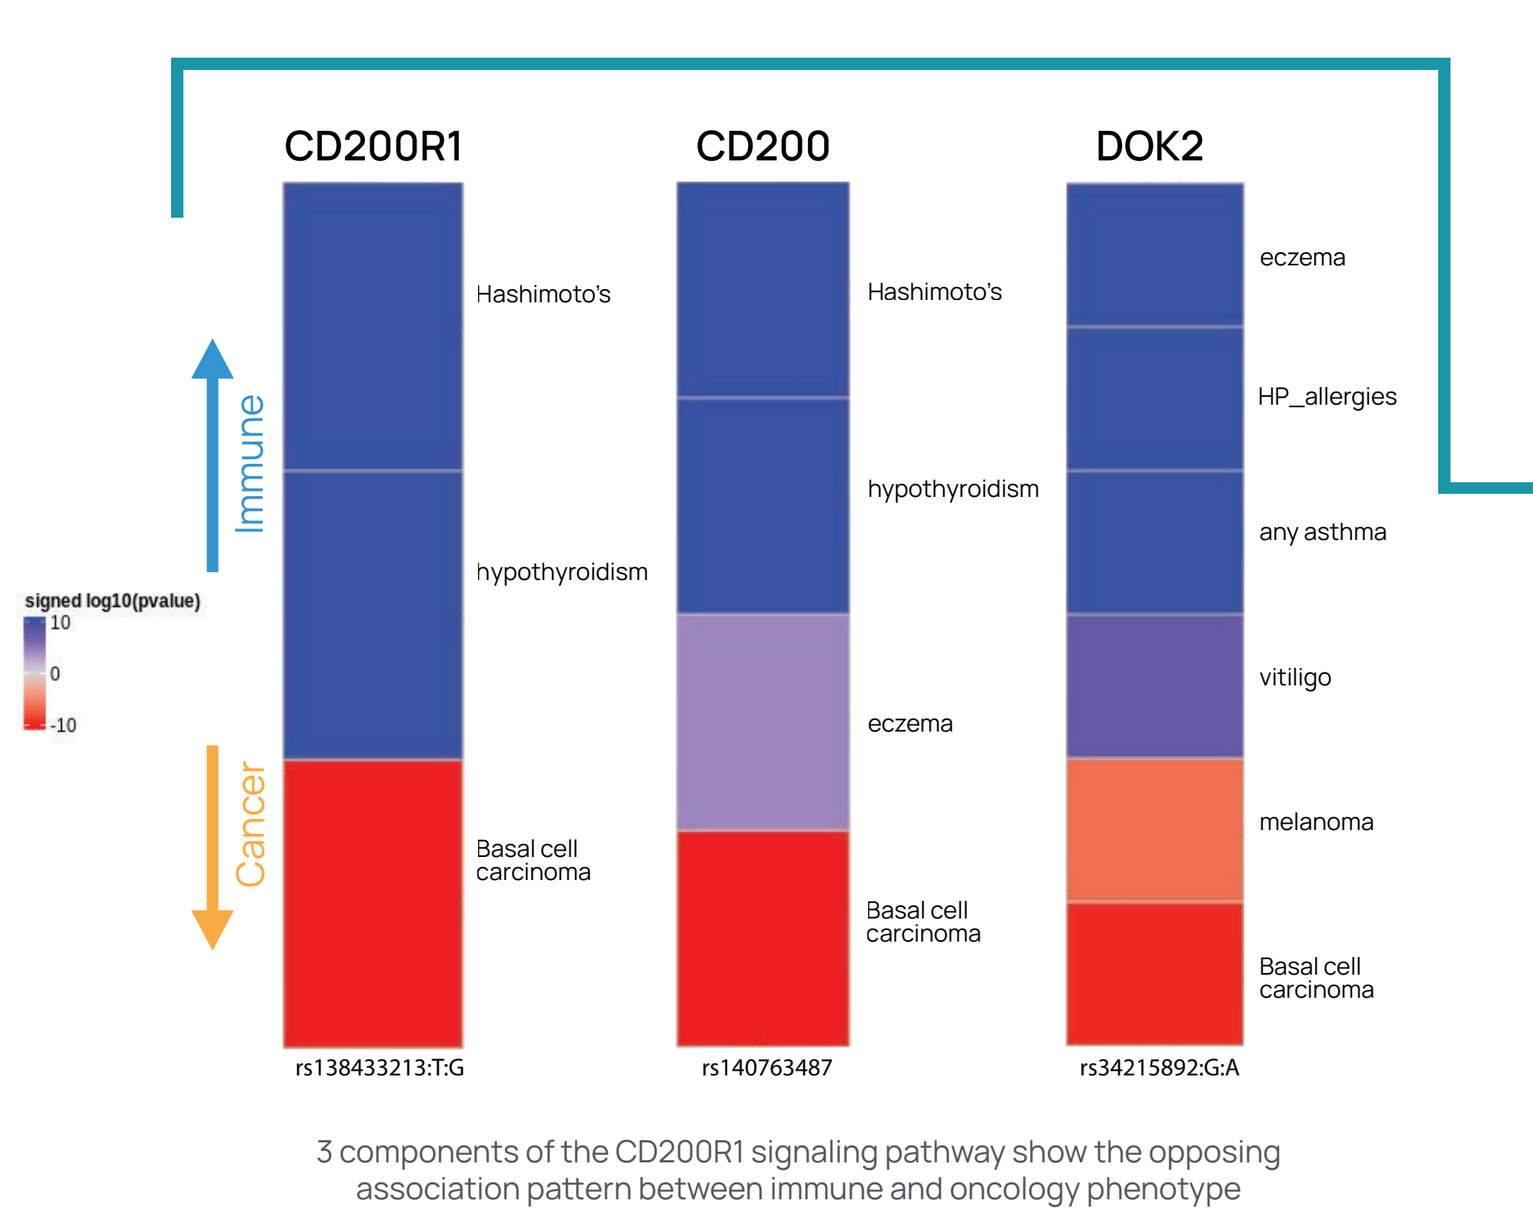

CD200R1
CD200
DOK2
Immune
Cancer
signed log10(pvalue)
Hashimoto's
hypothyroidism
Basal cell carcinoma
eczema
HP_allergies
any asthma
vitiligo
melanoma
rs138433213:T:G
rs140763487
rs34215892:G:A

3 components of the CD200R1 signaling pathway show the opposing association pattern between immune and oncology phenotype

I/O genetic signature shows opposing effects on autoimmune and cancer phenotypes

Figure 2: 23ME-00610 ('610), a Fully Humanized, Effectorless IgG1, Inhibits Immunosuppressive CD200/R1 Signaling via High Affinity Binding to CD200R1

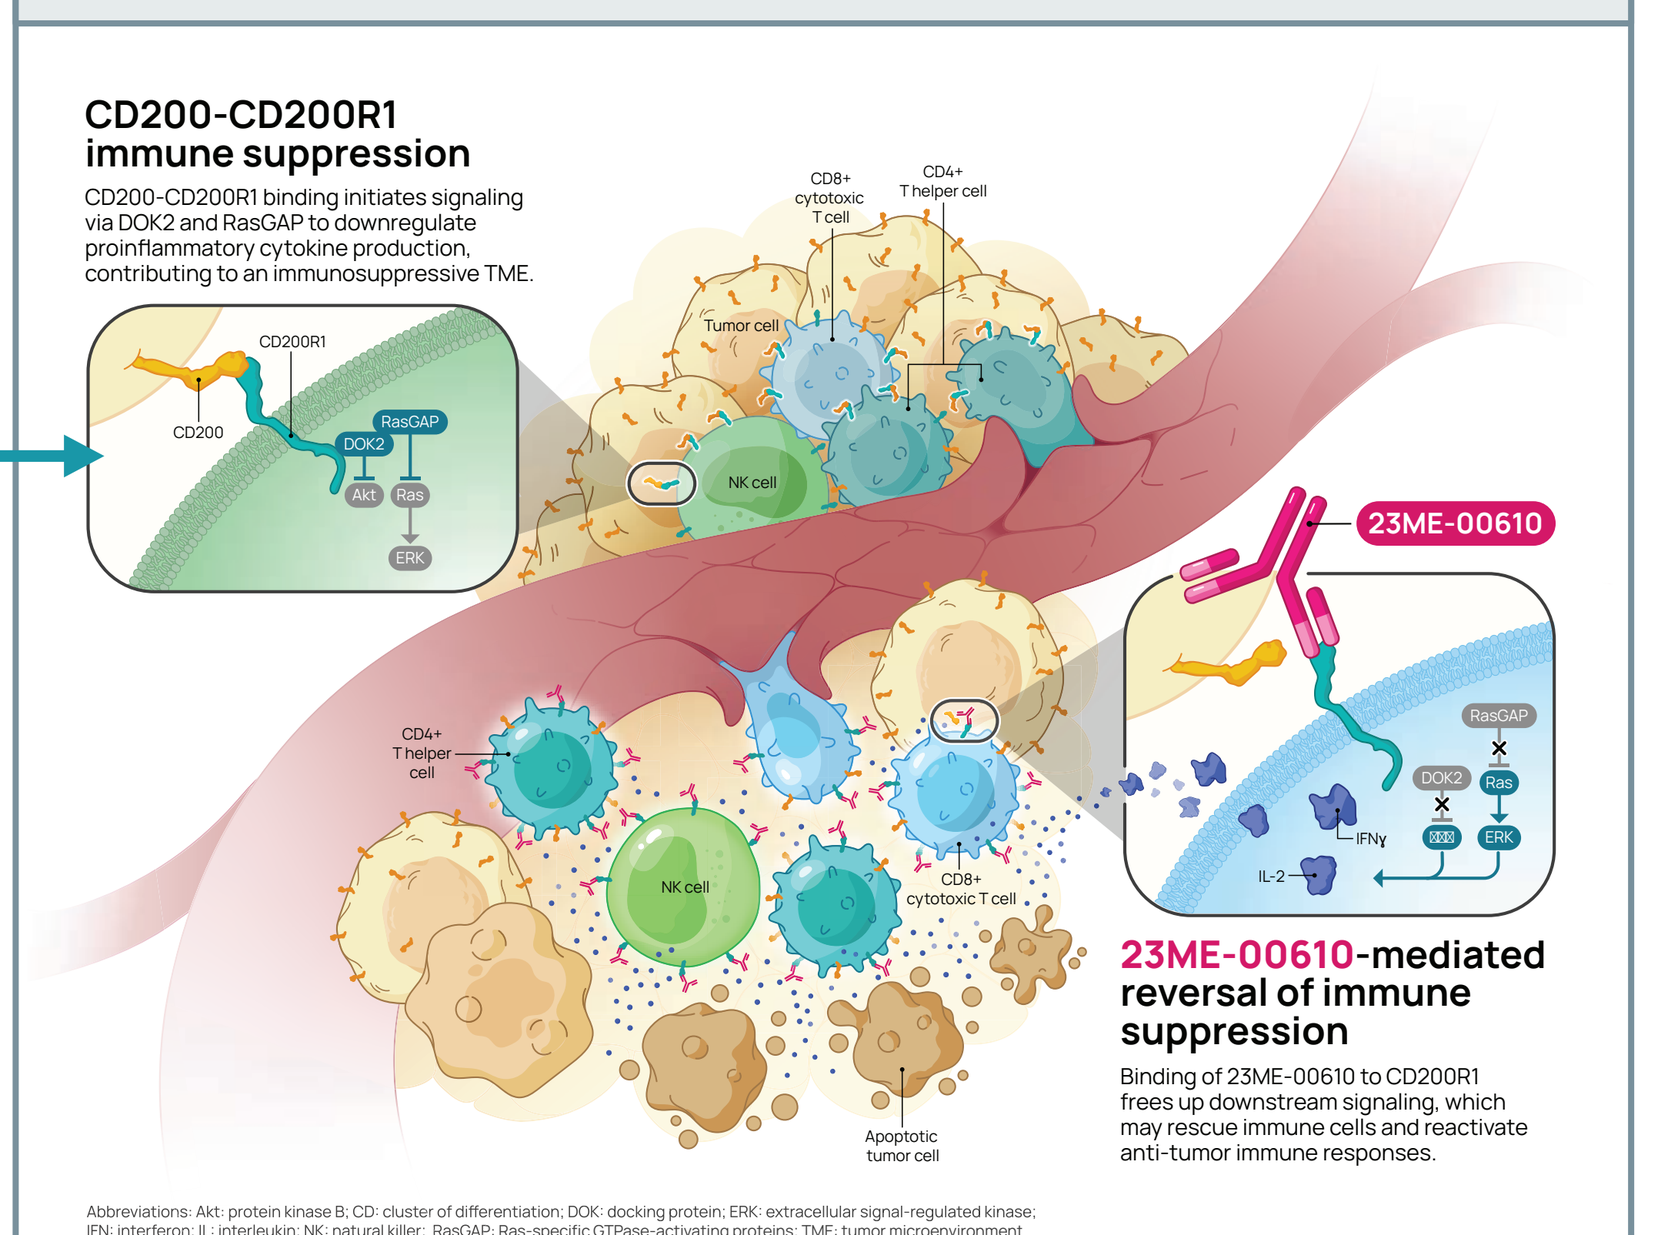

CD200-CD200R1 immune suppression
CD200-CD200R1 binding initiates signaling via DOK2 and RasGAP to downregulate proinflammatory cytokine production, contributing to an immunosuppressive TME.
23ME-00610
23ME-00610-mediated reversal of immune suppression
Binding of 23ME-00610 to CD200R1 frees up downstream signaling, which may rescue immune cells and reactivate anti-tumor immune responses.
NK cell
Tumor cell
Apoptotic tumor cell

Abbreviations: Akt: protein kinase B; CD: cluster of differentiation; DOK: docking protein; ERK: extracellular signal-regulated kinase; IFN: interferon; IL: interleukin; NK: natural killer; RasGAP: Ras-specific GTPase-activating proteins; TME: tumor microenvironment

METHODS

Figure 3: Phase 1/2 Study Design in Patients with Locally Advanced or Metastatic Solid Malignancies

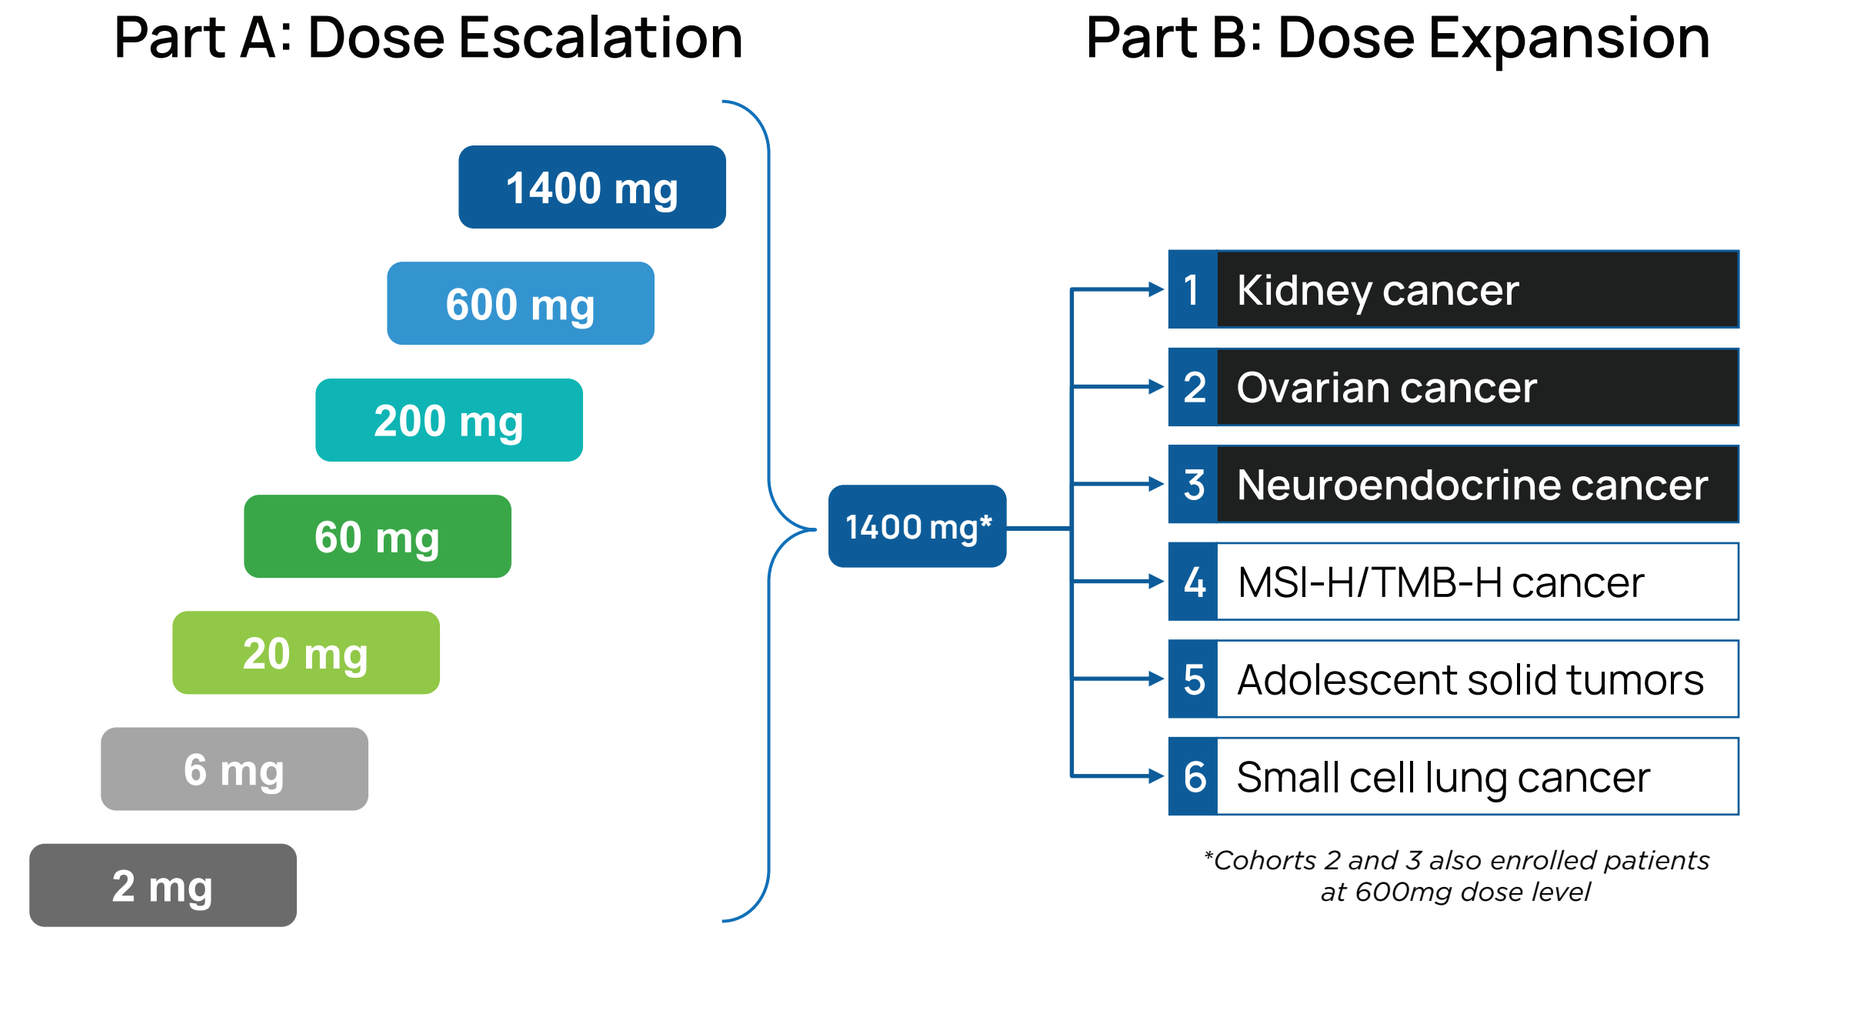

Part A: Dose Escalation
Part B: Dose Expansion
1400 mg
600 mg
200 mg
60 mg
20 mg
6 mg
2 mg
1400 mg*
1 Kidney cancer
2 Ovarian cancer
3 Neuroendocrine cancer
4 MSI-H/TMB-H cancer
5 Adolescent solid tumors
6 Small cell lung cancer
*Cohorts 2 and 3 also enrolled patients at 600mg dose level

Biomarker Assessments

Tumor Tissue Analysis

All Cohorts
- Assessment of pathway components
 - CD200R1 IHC (Ventana CD200R1 SP503 Assay)
 - CD200 IHC (Ventana CD200 SP517 Assay)

Neuroendocrine Cohort
- Based on early indications of anti-tumor efficacy (confirmed partial response), fresh tumor biopsies were collected pre- and on-treatment (Cycle 3 Day 1) to assess immune contexture at baseline and after 23ME-00610 treatment
 - multiplex IHC (CD3/8/68/163/GrB)
 - RNA seq

Neuroendocrine, Renal Cell Carcinoma and Ovarian Cancer Cohorts
- Orthogonal digital histopathology analysis on H&E-stained archival tumor sections using an AI-powered H&E analyzer (Lunit SCOPE IO platform)

Exploratory Genetic Analysis

Ancestry Corrected Polygenic Risk Score
- 23andMe Research Cohort2 data was used to construct ancestry specific polygenic risk scores (PRSs) for immune- and oncology-associated traits and applied to trial participants genotyped on the same 23andMe platform

- 23ME-0610 administered IV every 21 days
- Expansions had target enrollment of 15 patients per cohort

In biomarker assessments and correlation with efficacy of 23ME-00610, clinical benefit is defined as Partial Response (PR) + Stable Disease (SD) defined by RECIST criteria at first on treatment tumor assessment. No response is defined as tumor progression at the first tumor assessment.

RESULTS

Figure 4: Baseline tumor expression levels of CD200 may be associated with clinical benefit with 23ME-00610 treatment in neuroendocrine neoplasms and renal cell carcinomas.

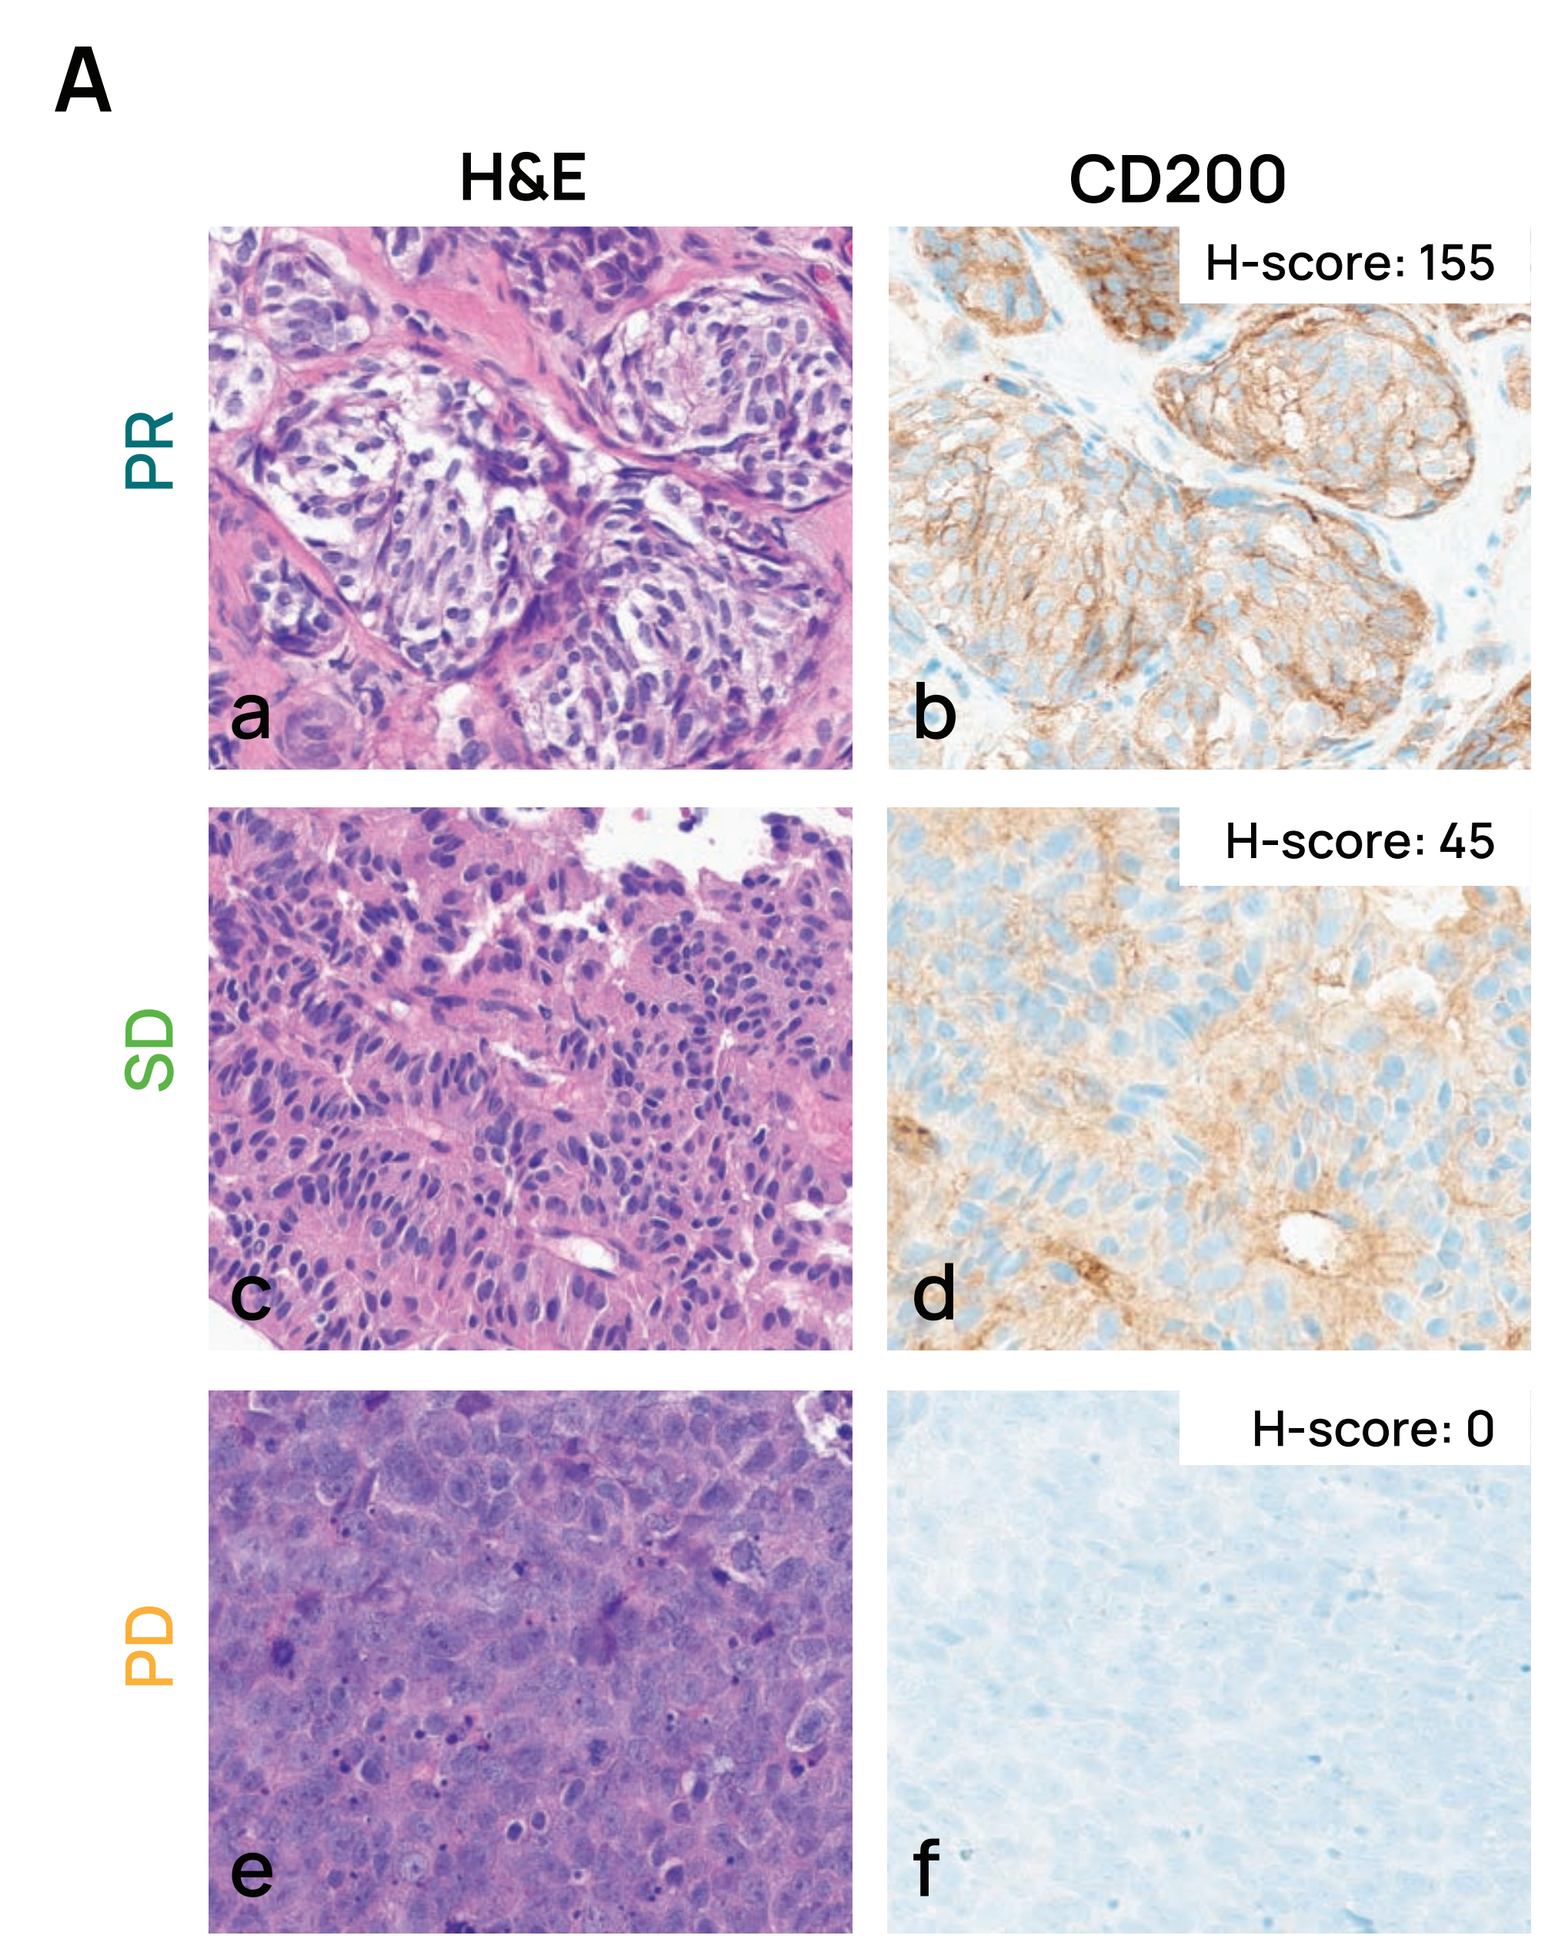

A
H&E
CD200
PR
SD
PD
H-score: 155
H-score: 45
H-score: 0
a
b
c
d
e
f

A. Representative images archival or fresh pre-treatment neuroendocrine neoplasms (NE) with high (b), moderate (d), and no CD200 expression (f) in the tumor cell membrane. Scale bar = 50 µm

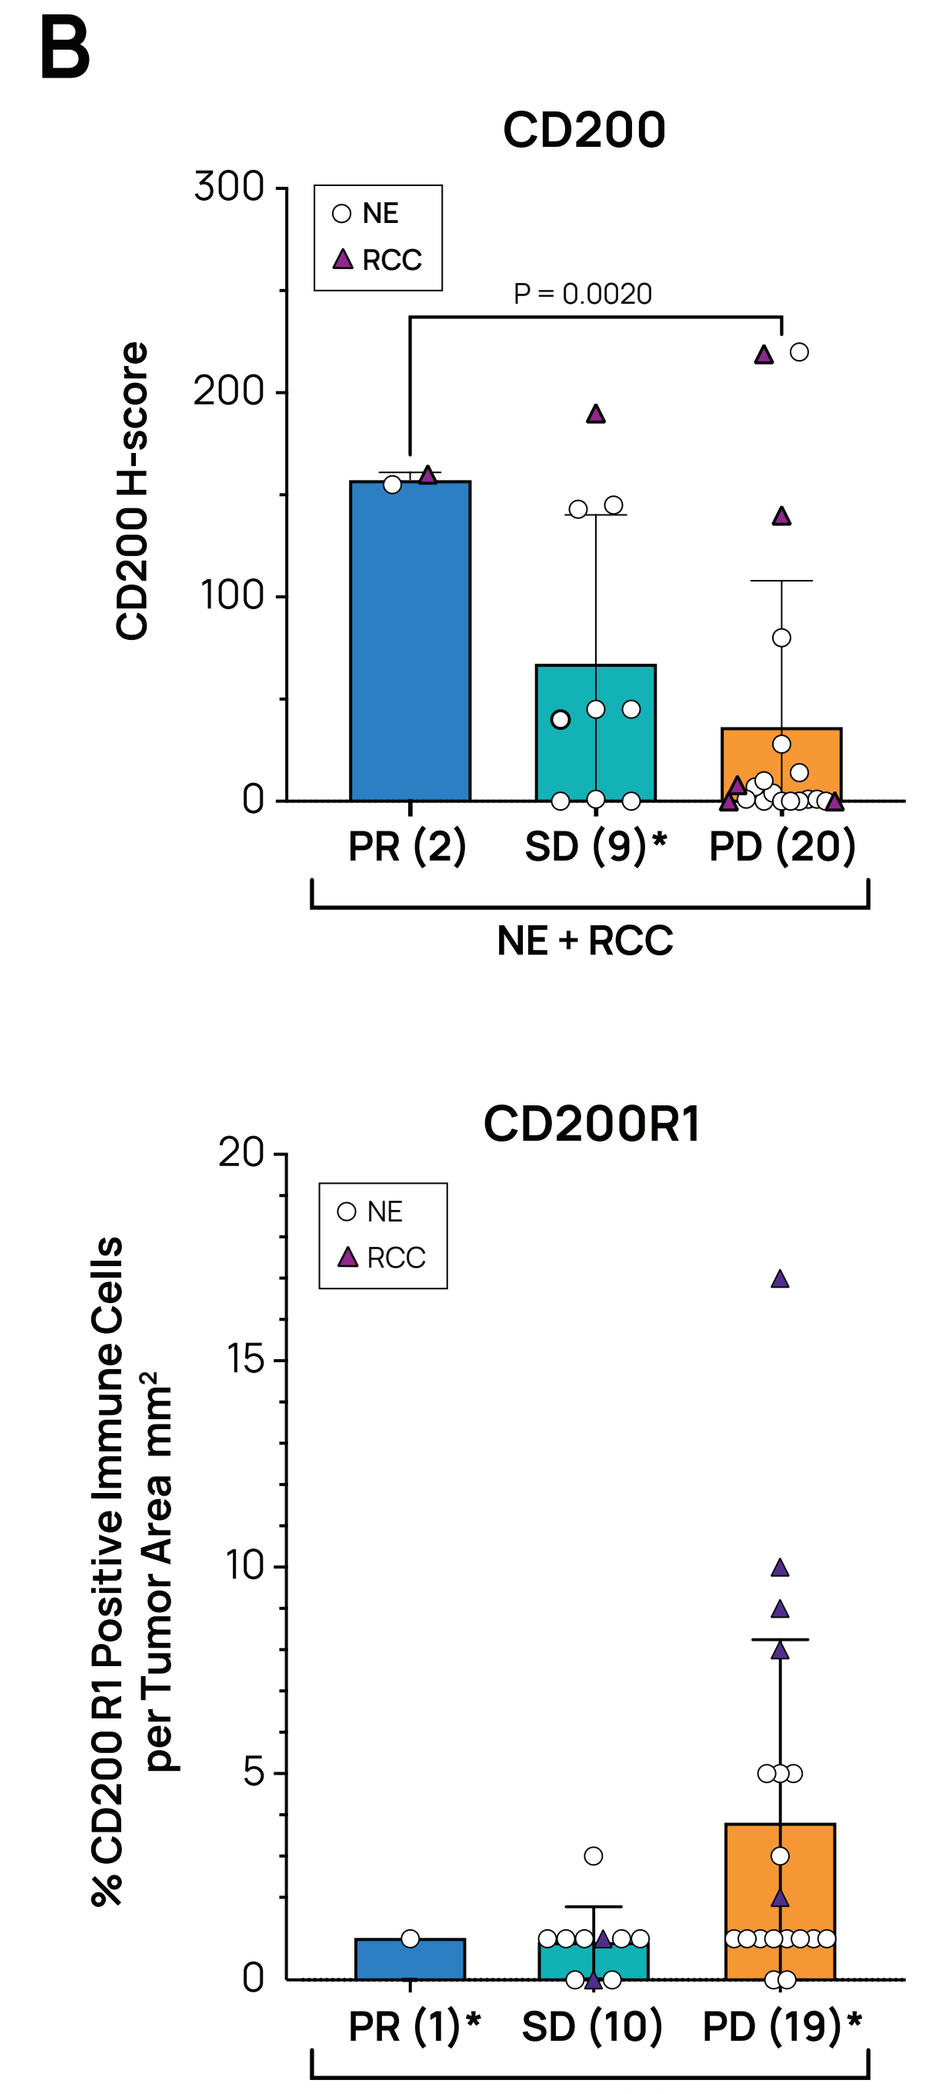

B
CD200
CD200 H-score
NE
RCC
P = 0.0020
PR (2)
SD (9)*
PD (20)
NE + RCC
CD200R1
% CD200 R1 Positive Immune Cells per Tumor Area mm²
PR (1)*
SD (10)
PD (19)*

B (top panel) High CD200 expression may be associated with clinical benefit on 23ME-00610 treatment in RCC and NE tumors (partial response (PR, n = 2, mean H-score = 157.5, st. dev = 3.5) or stable disease (SD, n = 9, mean H-score = 67.6, st. dev = 72.5). Patients that progressed (PD) on treatment on their first evaluation tended to have low to no CD200 expression (n = 20, mean H-score = 38.6, st. dev = 72.8). NE = neuroendocrine, RCC = renal cell carcinoma.

*1 patient did not have archival tissue that was evaluable or sufficient for IHC staining.

B (bottom panel) Expression of CD200R1 in tumor infiltrating immune cells was low to moderate in tumors of trial participants. It was not significantly associated with clinical response: PD (mean = 4%. st.dev = 4.6) vs. patients that derived some clinical benefit (PR or SD) (mean = 1%, st.dev = 0.9. CD200R1 expression data was not available for 8 patients due to lack of sufficient archival tissue.

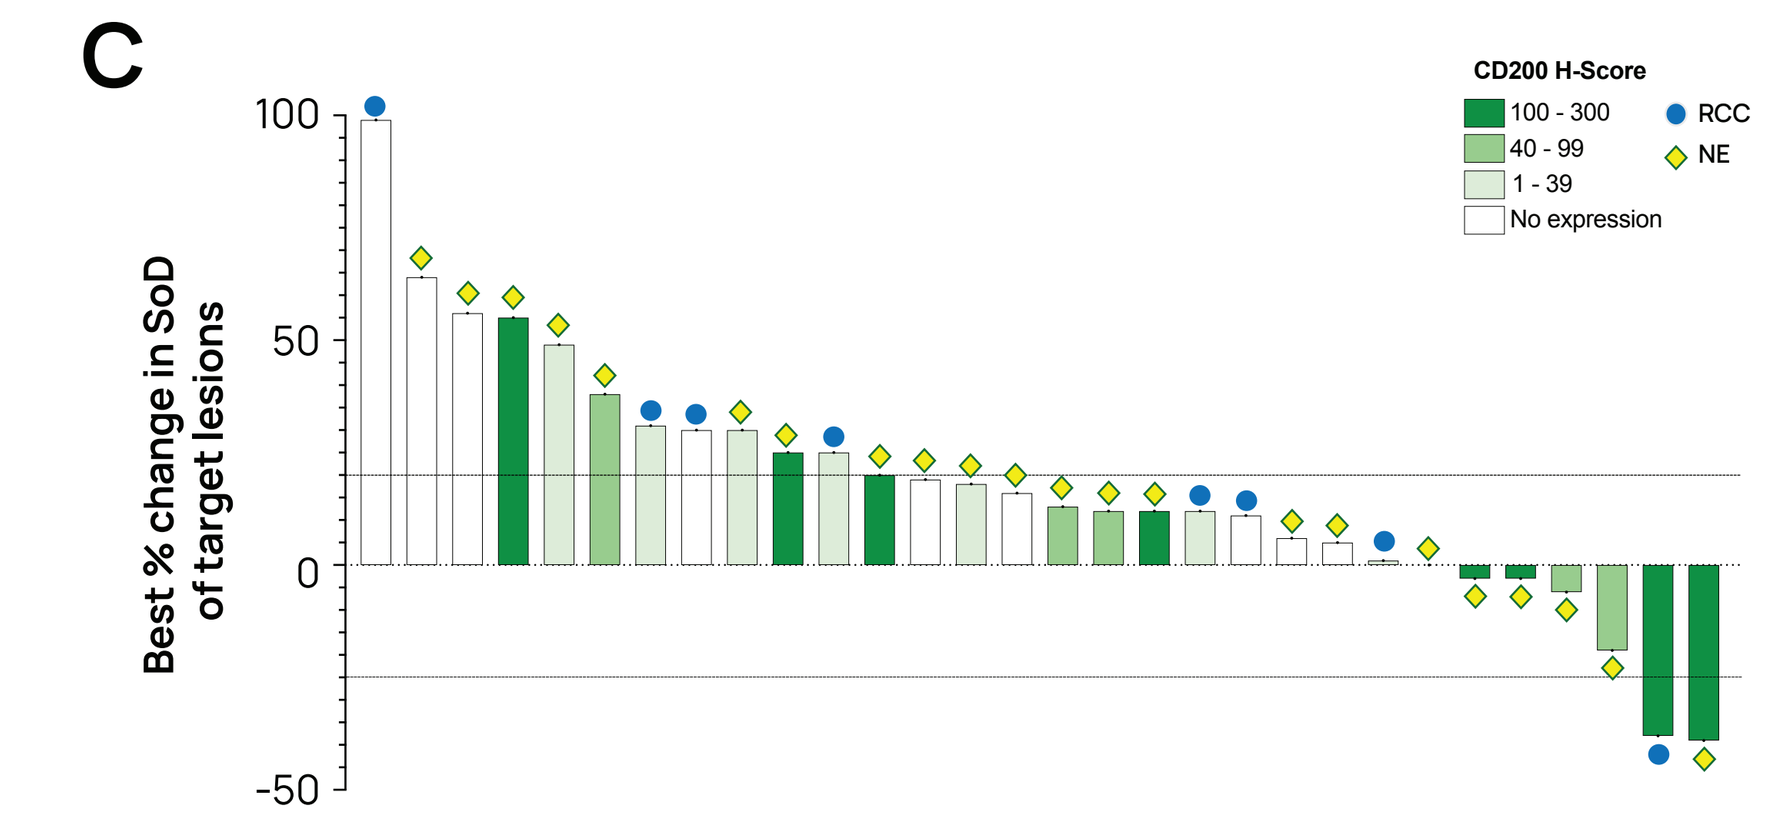

C
Best % change in SoD of target lesions
CD200 H-Score
100 - 300
40 - 99
1 - 39
No expression
RCC
NE

C. RCC and NE tumors that had the best summary of target lesion diameters tended to have higher CD200 expression.

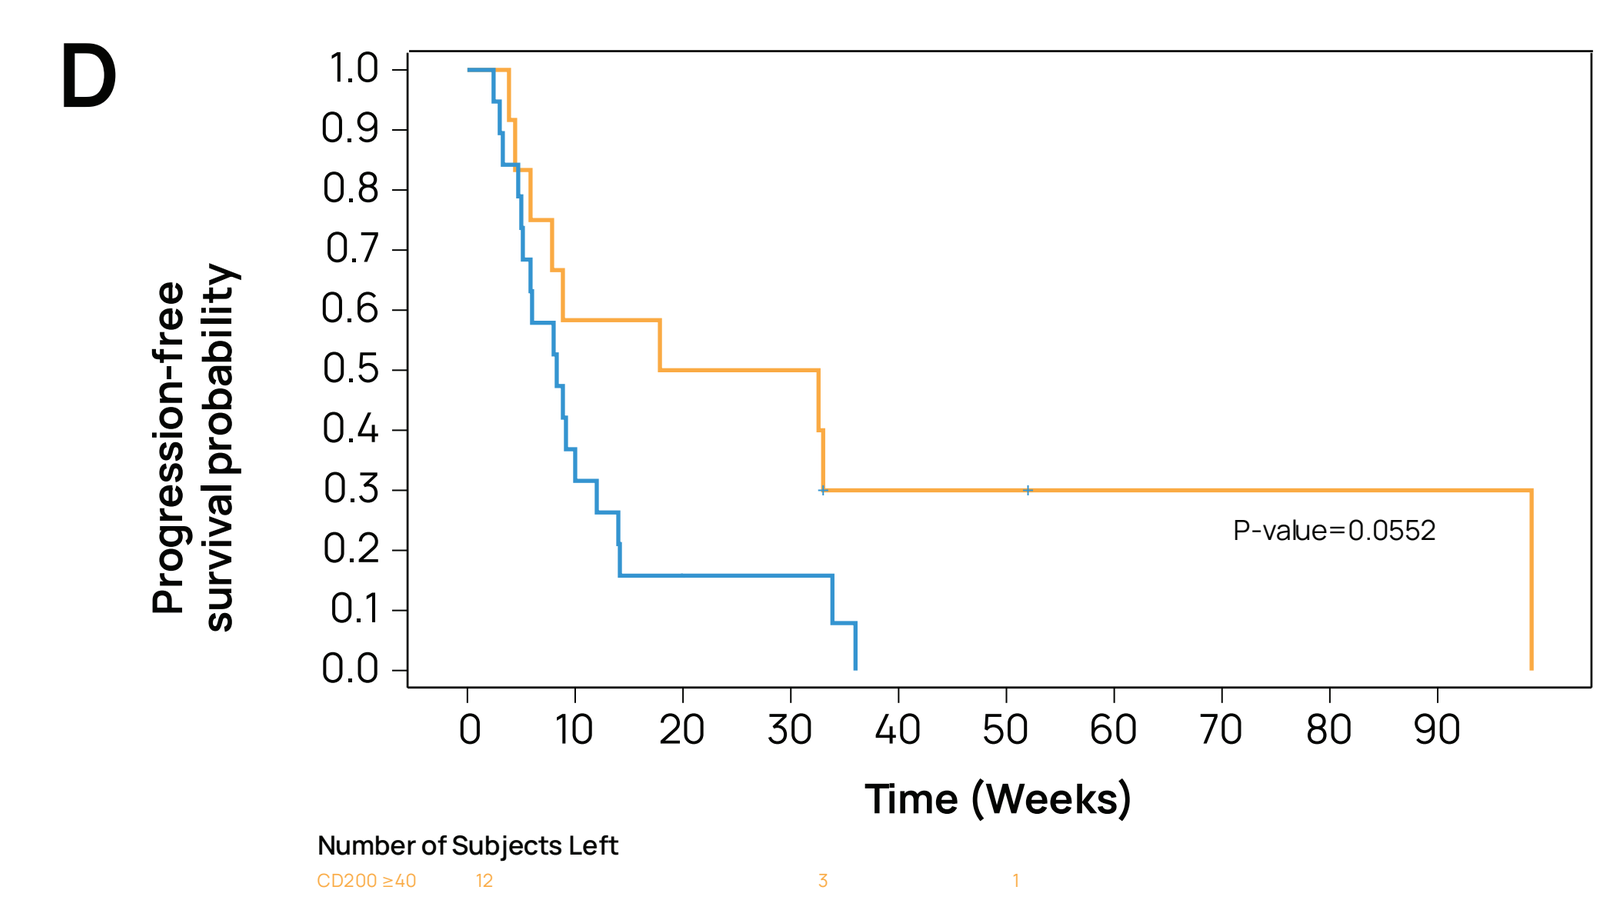

D
Progression-free survival probability
Time (Weeks)
P-value=0.0552
Number of Subjects Left

D. Progression free survival of NE and RCC patients: trial participants with tumor CD200 H-scores >40 tended to have longer progression free survival than those with CD200 H-scores <40. Univariate (time-to-event) log rank test used for statistical analysis.

Figure 5. Host Genetics: Polygenic Risk Scores May Be Leveraged to Augment the Association of Tumor CD200 Expression With Clinical Benefit (PR/SD) of 23ME-00610 Treatment

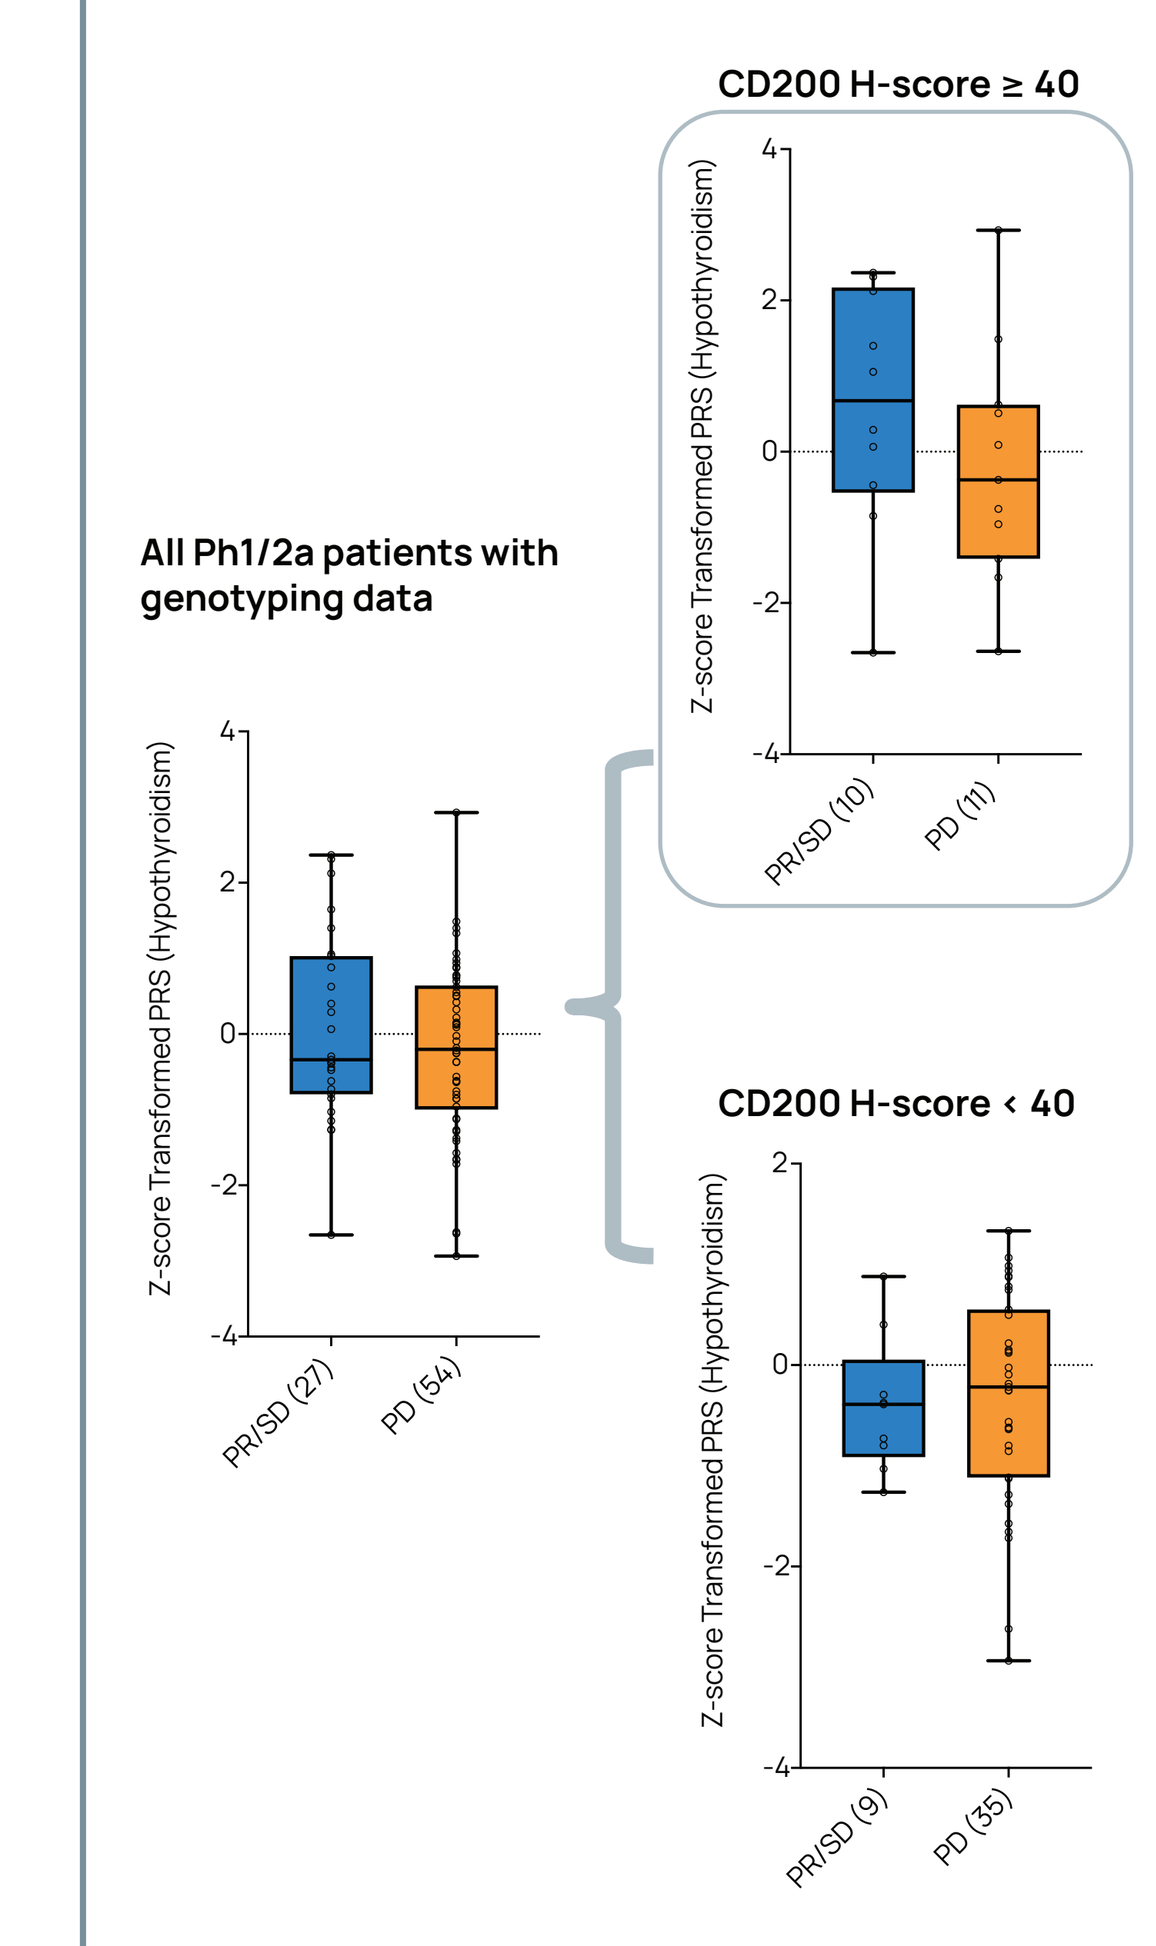

All Ph1/2a patients with genotyping data
CD200 H-score ≥ 40
CD200 H-score < 40
Z-score Transformed PRS (Hypothyroidism)
PR/SD (27)
PD (54)
PR/SD (10)
PD (11)
PR/SD (9)
PD (35)

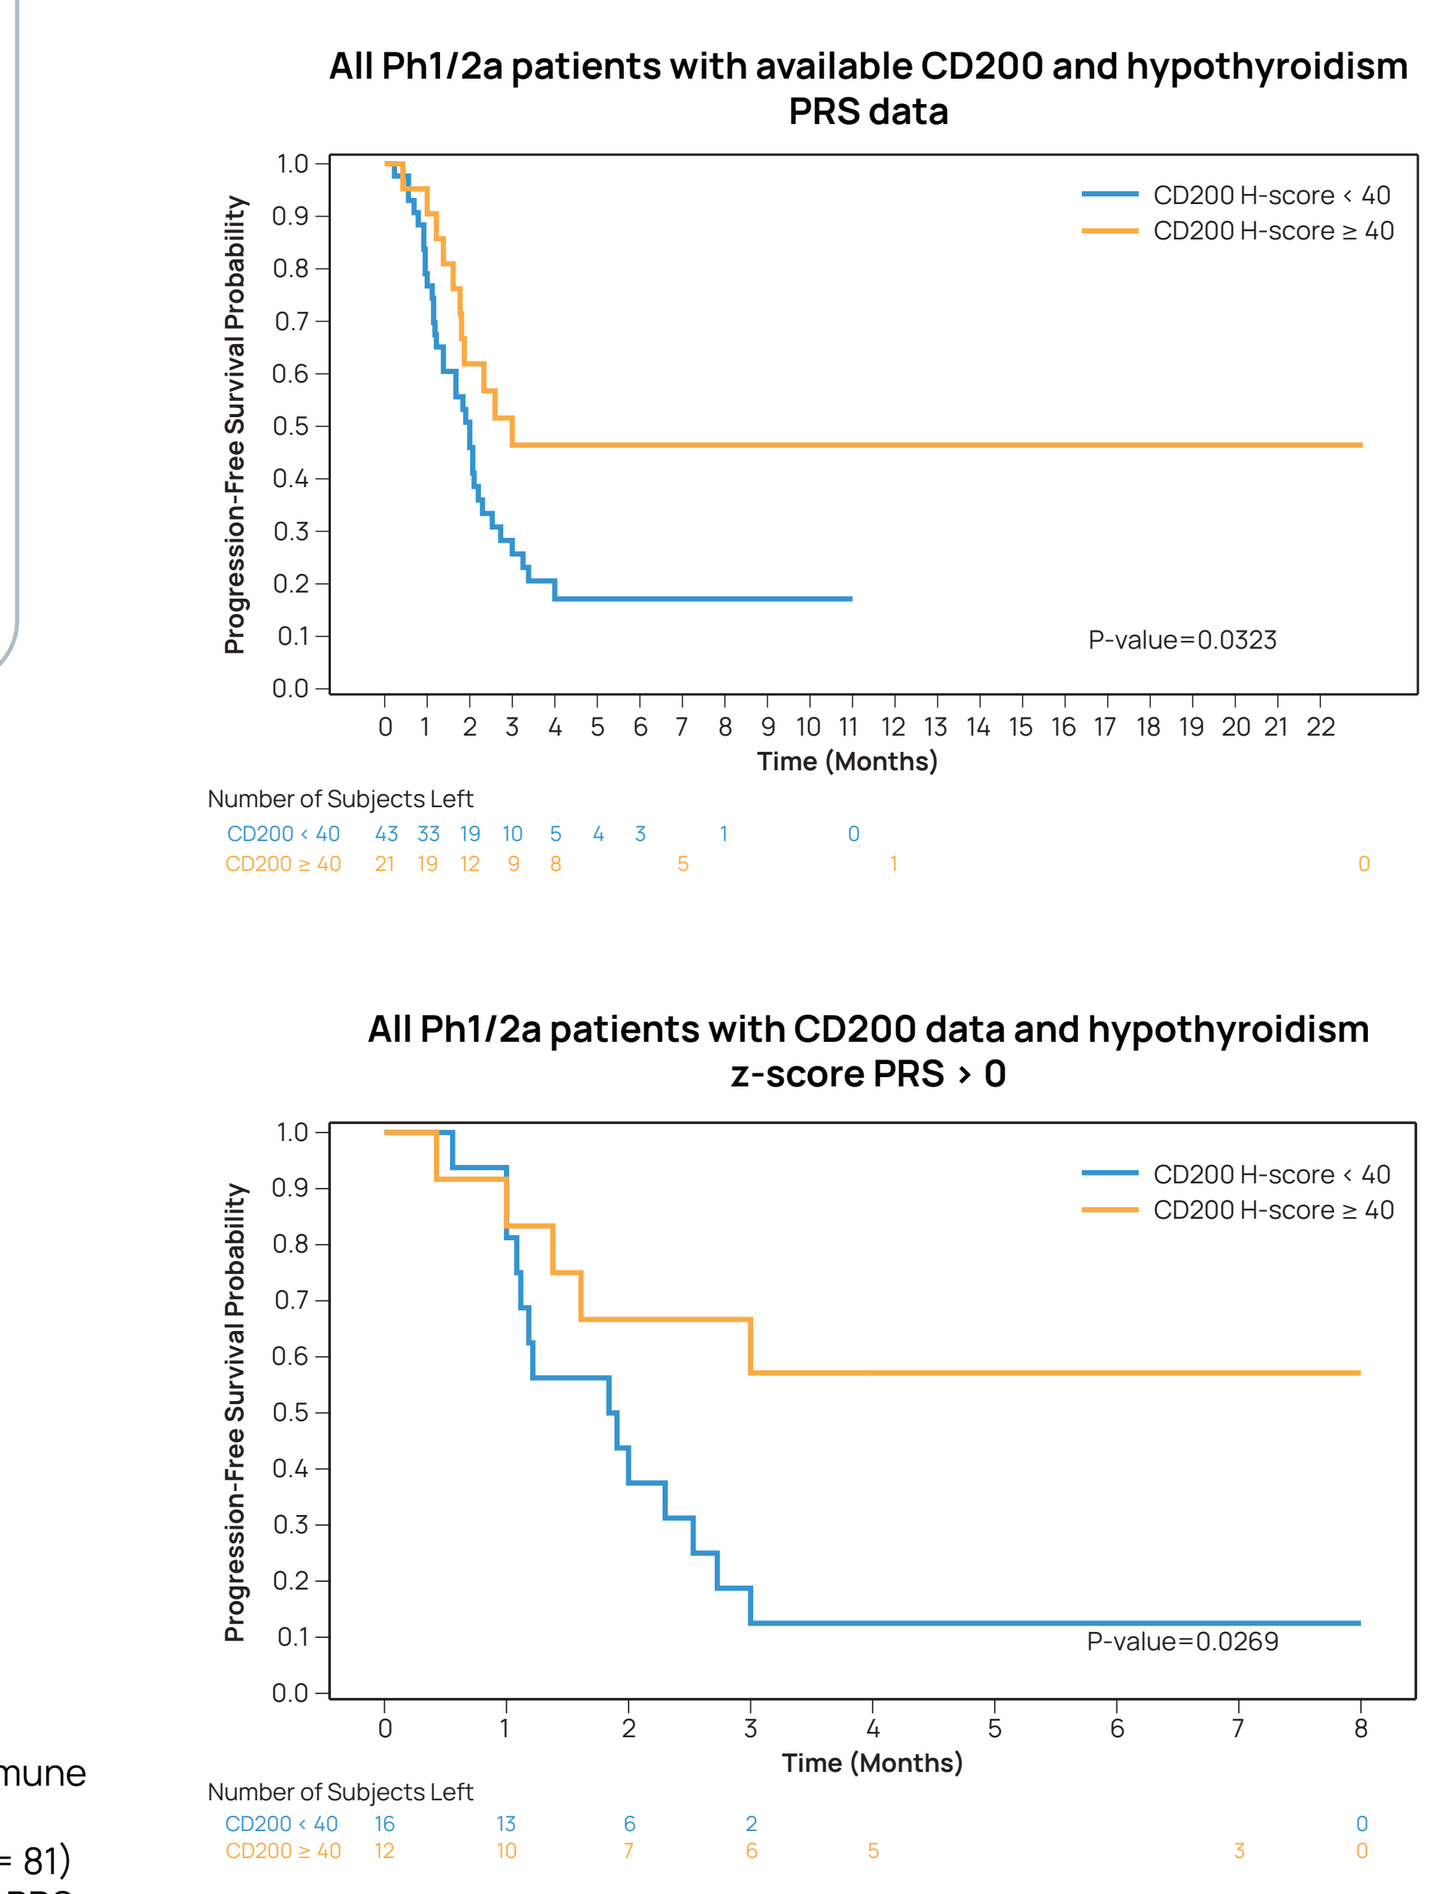

All Ph1/2a patients with available CD200 and hypothyroidism PRS data
Progression-Free Survival Probability
CD200 H-score < 40
CD200 H-score ≥ 40
P-value=0.0323
Time (Months)
Number of Subjects Left
All Ph1/2a patients with CD200 data and hypothyroidism z-score PRS > 0
P-value=0.0269

- To assess the potential interaction of genetic host immune and tumor parameters on 23ME-00610 efficacy, all Phase 1/2a enrolled patients with genotyping data (n = 81) had ancestry corrected immune-mediated phenotype PRS calculated. The polygenic risk score (PRS) was trained on the research consented 23andME customer database.
- For the hypothyroidism phenotype, there is no difference in median PRS (Z-score transformed using healthy ancestry matched population means), between patients that had a PR or SD vs. PD.
- When segregated based on "high" CD200 tumor expression (H-score > 40), patients with high CD200 who achieved a PR or SD had a higher median (0.6728, ICR = 5.020) genetic risk for hypothyroidism than patients with high CD200 who had PD (median = -0.3707, ICR = 5.568).

- The combination of CD200 high expression and hypothyroidism PRS > 0 may have a better predictive association with longer time to progression. Univariate (time-to-event) analysis using a log rank test was performed.
- This data might indicate that the immune host genetics might influence the ability of a patient to raise a sufficient anti-tumor immune response against CD200 expression tumors and benefit from CD200R1 blockade.

Figure 6: Baseline Tumor Immune Contexture: Clinical Benefit May Be More Likely In Immune Low Though More Immune Permissive Tumor Micro-environments (Macrophages < Lymphocytes)

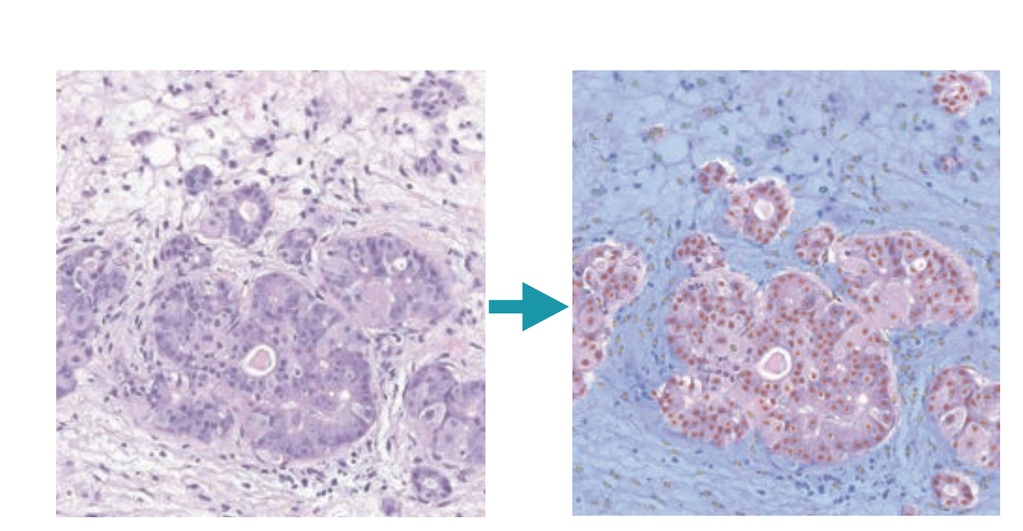

A

Tumor cells: orange
Blue mask: stroma
Lymphocytes: yellow
Macrophages: green

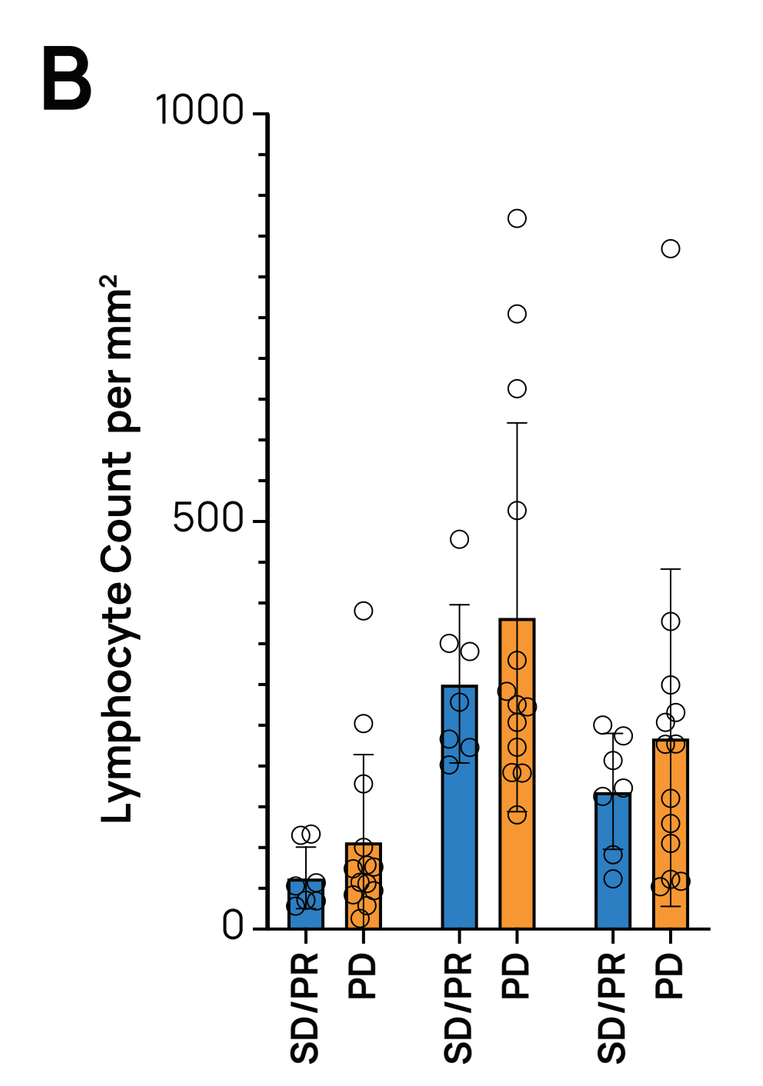

B
Lymphocyte Count per mm²
SD/PR
PD
Cancer
Stroma
Combined

A. AI powered algorithm was applied to H&E images of archival or pre-treatment fresh biopsies from the neuroendocrine cohort to determine spatially defined immune infiltrates (Lunit SCOPE IO).

Tumors characterized by immune low or excluded phenotype might be more likely to benefit from 23ME-00610 treatment: Tumor infiltrating lymphocytes **(B)**, and macrophages **(C)** were quantified in tumor area, tumor stromal area and combined total tumor and stromal areas. Tumors from patients deriving clinical benefit (PR, SD) during 23ME-00610 treatment had fewer infiltrating lymphocytes and macrophages when compared to tumors that progressed at the first radiological tumor assessment (PD).

More immune permissive tumor micro-environments might be more likely to lead to benefit from 23ME-00610: Macrophage to lymphocyte cell density ratios were also calculated **(D)**, with relatively lower macrophage infiltration in relation to lymphocytes associating with clinical benefit.

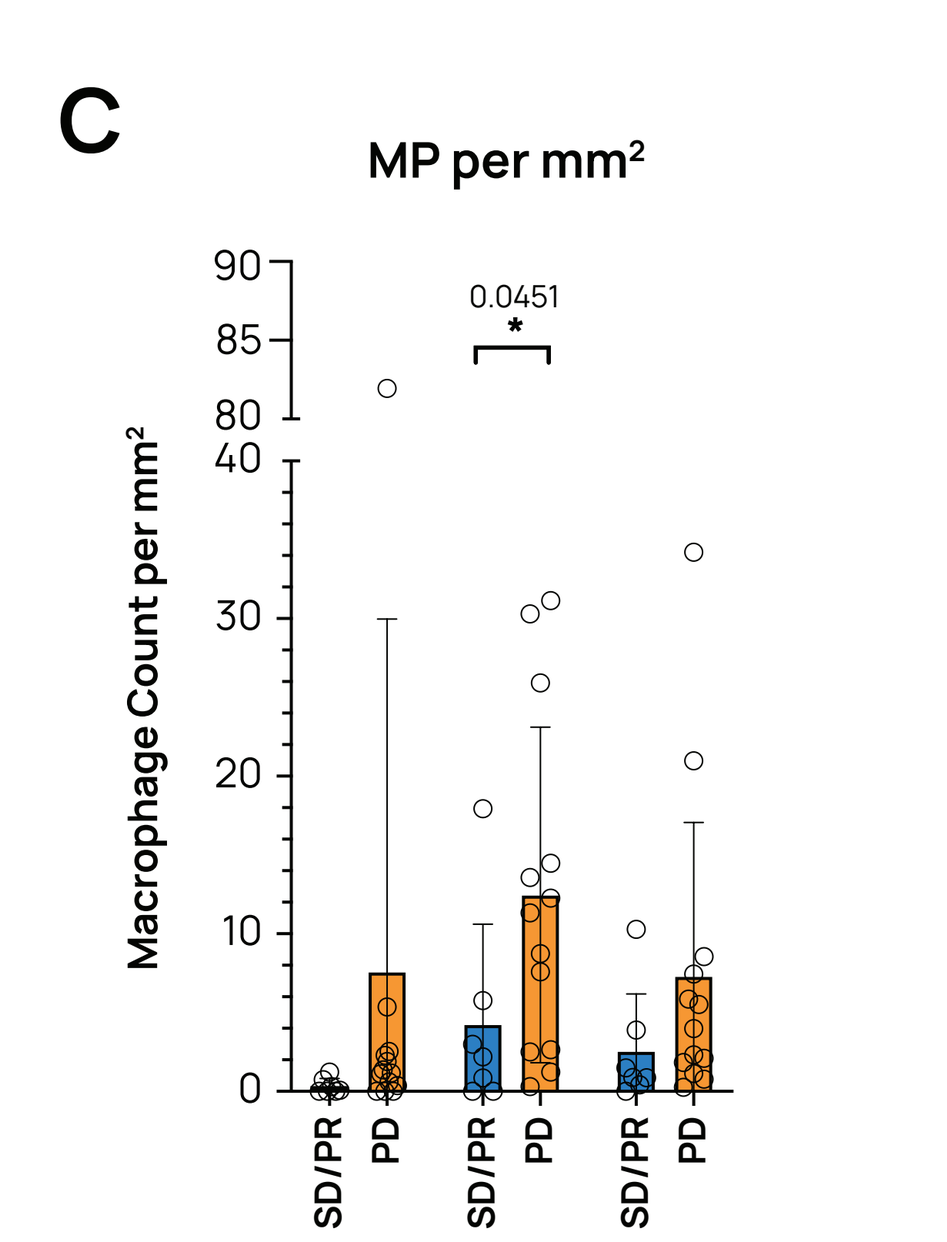

C
MP per mm²
Macrophage Count per mm²
0.0451
SD/PR
PD
Cancer
Stroma
Combined

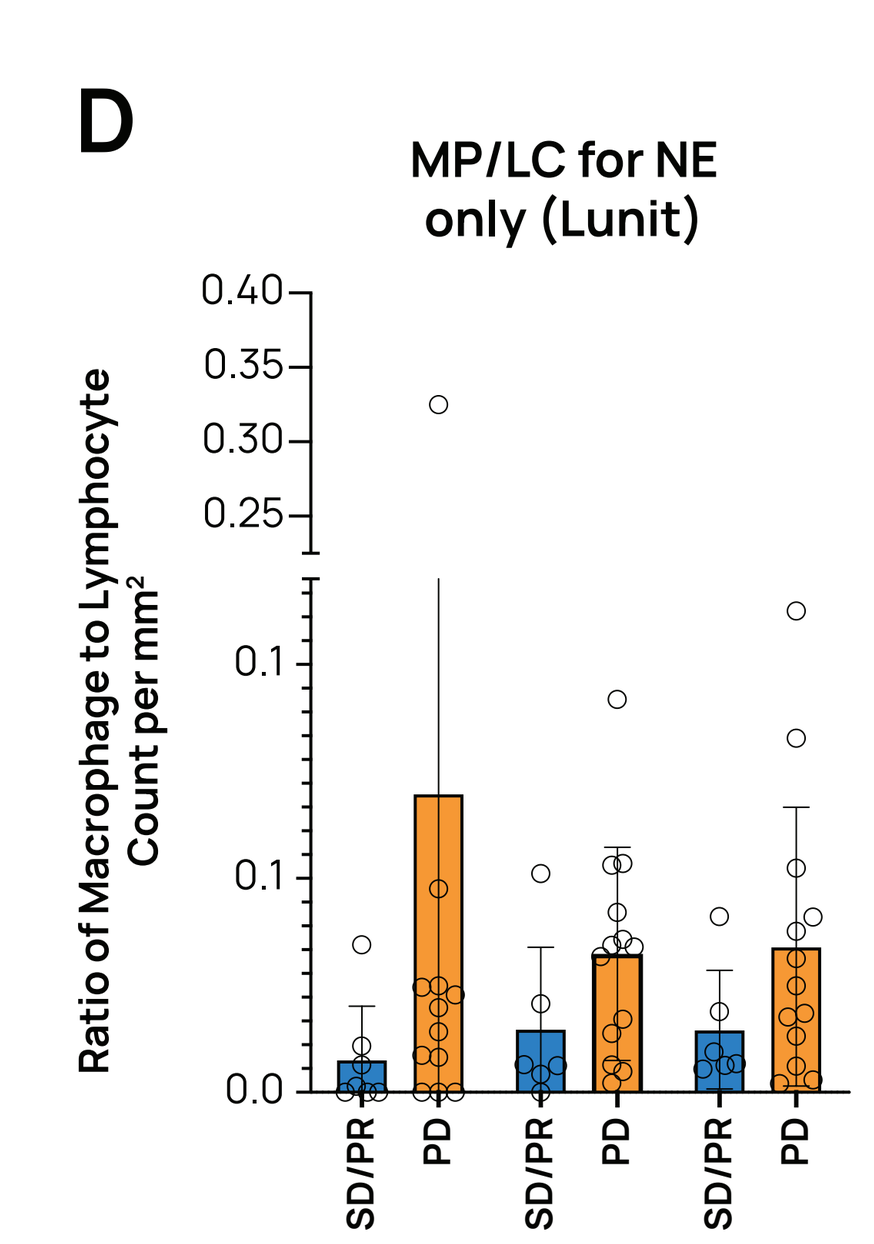

D
MP/LC for NE only (Lunit)
Ratio of Macrophage to Lymphocyte Count per mm²
SD/PR
PD
Cancer
Stroma
Combined

Figure 7. Pharmacodynamics: 23ME-00610 Leads to Changes in the Immune Composition and Immune Activation

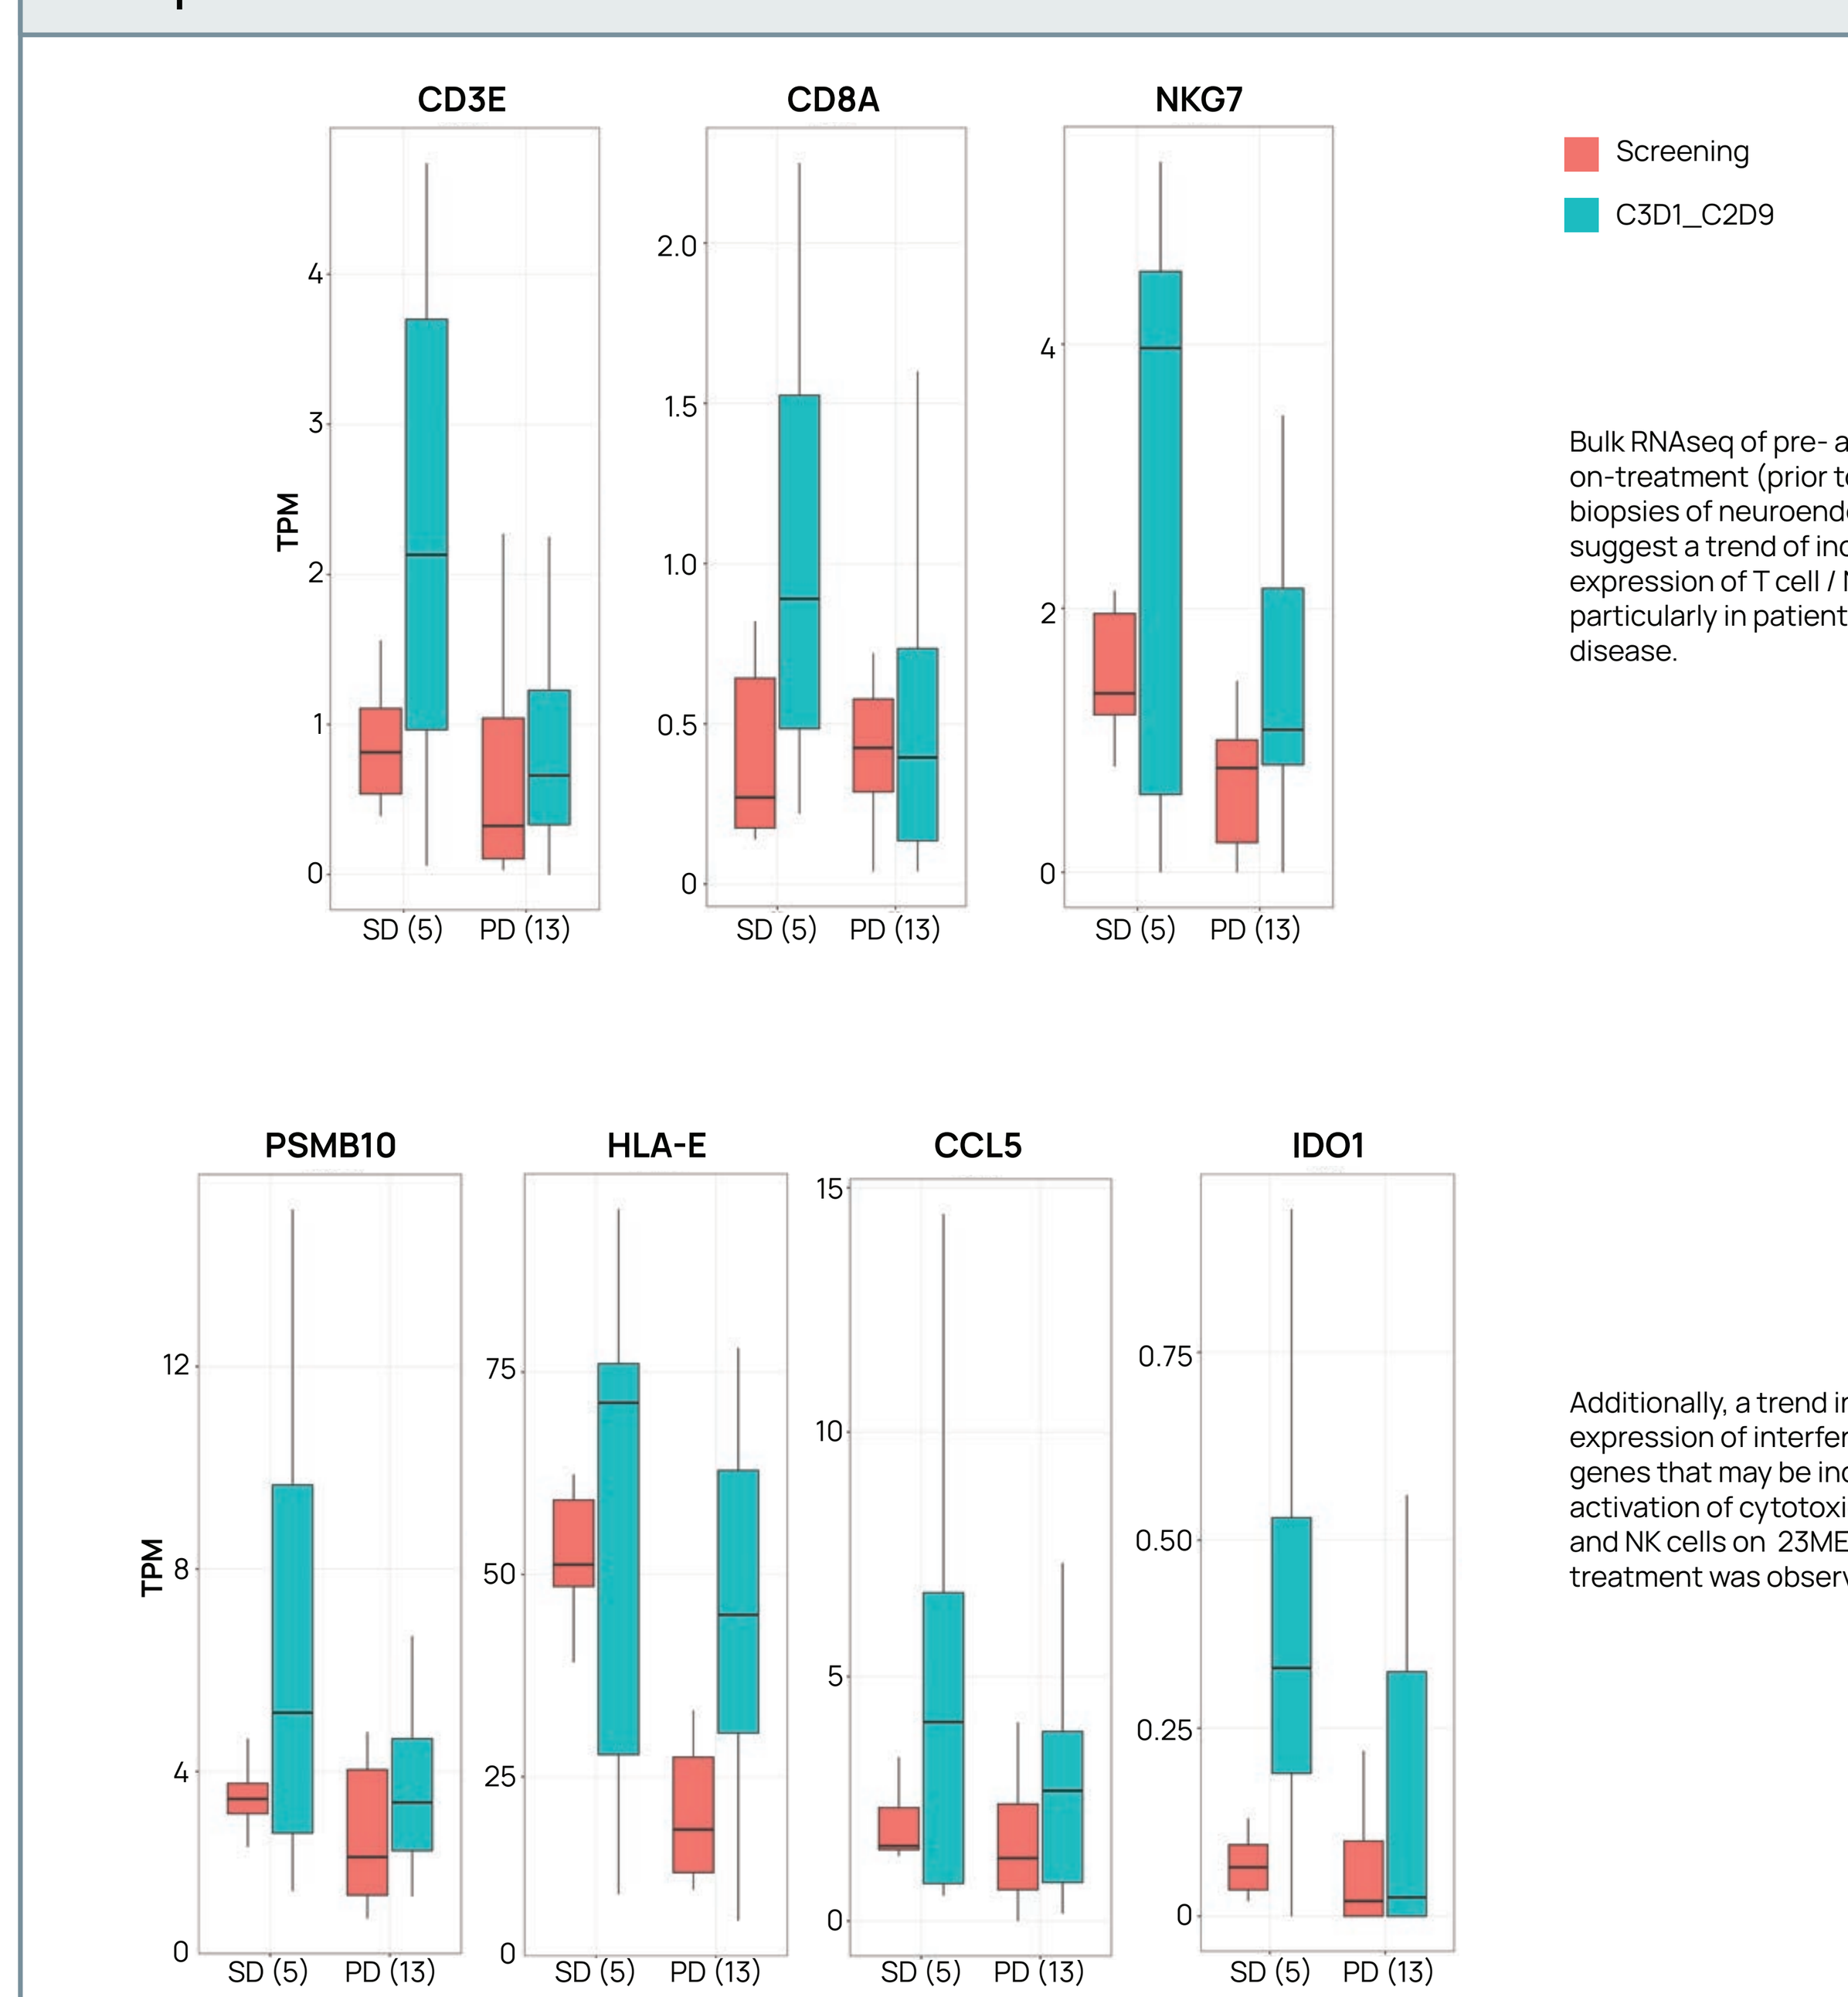

CD3E
CD8A
NKG7
PSMB10
HLA-E
CCL5
IDO1
TPM
SD (5)
PD (13)
Screening
C3D1_C2D9

Bulk RNAseq of pre- and on-treatment (prior to Cycle 3) tumor biopsies of neuroendocrine tumors suggest a trend of increased gene expression of T cell / NK cell markers, particularly in patients with stable disease.

Additionally, a trend in increased expression of interferon inducible genes that may be indicative of activation of cytotoxic CD8+ T cells and NK cells on 23ME-00610 treatment was observed .

CONCLUSIONS

- Higher tumor cell expression of CD200 associates with clinical benefit from 23ME-00610 in some patients, and this tumor based putative biomarker might be further augmented if combined with genetic based host immune set point readouts (e.g. hypothyroidism PRS), warranting further exploration of this and other biomarkers for potential future patient selection.
- Neuroendocrine tumors that had tumor shrinkage or prolonged stable disease tended to be less inflamed at baseline potentially due to CD200 mediated immune suppression/exclusion, though these tumors might be more permissive to immune activation due to lower macrophage to lymphocyte ratio.
- Analysis of pre- and on-treatment tumor samples showed an increase in T and NK cell markers and an increase in interferon inducible genes with 23ME-00610 treatment suggesting pharmacodynamic immune modulation.

REFERENCES

1. Fenaux J, et al. *Oncoimmunology*. 2023; 2. Mihrshahi R, et al. *J Immunol*. 2009; 3. Timmerman LM, et al. *PLoS One*. 2021; 4. Misstear K, et al. *J Virol*. 2012; 5. Salek-Ardakani S, et al. *Eur J Immunol*. 2019; 6. Choueiry F, et al. *J Immunother Cancer*. 2020; 7. Moreaux J, et al. *Biochem Biophys Res Commun*. 2008; 8. Vathiotis IA, et al. *Cancers (Basel)*. 2021; 9. Love JE, et al. *Am J Clin Pathol*. 2017; 10. Kummar S, et al. *Cancer Res*. 2023; 11. Rasco D, et al. *J Immunotherap Cancer*. 2023; 12. Glatt DM, et al. *Journal for ImmunoTherapy of Cancer*. 2023; 13. Khan Z, et al. *Proc Natl Acad Sci USA*. 2020.

Study sponsored by 23andMe, Inc.

Corresponding author email address: Albiruni.Razak@uhn.ca

DOI for presenting author, Albiruni Razak: Consulting or Advisory Role - Adaptimmune; GlaxoSmithKline; Inhibrx; Medison Research Funding - 23 and Me; Abbvie; Adaptimmune; Amgen; Blueprint Medicines; Boehringer Ingelheim; Bristol-Myers Squibb; Daiichi Sankyo; Deciphera; Frontier Therapeutics; GlaxoSmithKline; Iterion Therapeutics; Karyopharm Therapeutics; MedImmune; Merck; Neoleukin Therapeutics; Pfizer; Rain Therapeutics; Roche/Genentech; Symphogen; Expert Testimony - Medison